Pharmacologic inhibition of FASN reverses diet-induced steatohepatitis in mice and TVB-2640 inhibits lipogenesis in humans

Elizabeth J. Parks [2,3], Camila M. Manrique[4], Majid M. Syed-Abdul[2], Ayman H. Gaballah[5], Ghassan M. Hammoud[3], Douglas Buckley[1], Greg Duke[1], William McCulloch[1], George Kemble[1]

[1] 3-V Biosciences, Inc. Menlo Park, CA; [2] Department of Nutrition and Exercise Physiology; [3] Division of Gastroenterology and Hepatology, Univ. of Missouri, Columbia, MO; [4] Division of Endocrinology, Univ. of Missouri School of Medicine; and [5]Department of Radiology, Univ. of Missouri School of Medicine, Columbia, MO.

AASLD THE LIVER MEETING® 2017 WASHINGTON, DC OCTOBER 20-24

INTRODUCTION

Dietary sugars induce an increase in hepatic de novo lipogenesis (DNL), which left unchecked promotes liver inflammation, ultimately leading to the development of fibrosis and nonalcoholic steatohepatitis (NASH). Pharmacologic inhibition of fatty acid synthase (FASN), a key enzyme in the DNL pathway, treats steatosis, inflammation and fibrosis in high-fat, high-fructose fed (HFFD) murine models (see **Poster 1994** for a full description of the murine data).

One compelling mechanism for NASH treatment is inhibition of the driver of this progressive disease, specifically reduction of hepatic lipogenesis mediated by FASN. 3-V Biosciences has developed the highly selective FASN inhibitor, TVB-2640, and evaluated it in early phase clinical studies.

AIM

The primary objective of this human pharmacology study was to determine the extent that TVB-2640 could inhibit DNL in subjects with BMI >26 and characteristics of metabolic syndrome.

MATERIAL & METHODS

For murine studies, animals were fed high fat, high fructose diet (HFFD) for 44 weeks and the presence of liver fibrosis was confirmed by biopsy. The HFFD diet was continued for an additional 8 weeks in addition to daily oral dosing with the FASN inhibitor, TVB-3664, or vehicle without drug (Fig. 1). For a full description of the methods used for the murine model, please see Poster 1994.

The PK of TVB-2640 was evaluated in 20 healthy volunteers; each subject received one 50mg capsule of drug on an empty stomach and blood collected at intervals up to 48 hours (Fig. 2).

The impact of TVB-2640 on DNL was evaluated in healthy males with a BMI above 26 and characteristics of metabolic syndrome (ie elevated triglycerides and fasting blood glucose levels) (Table 1). DNL was measured once prior to dosing and once after 10 days of once-daily, oral TVB-2640 (Fig. 3). To measure DNL, the subject received an IV infusion of $^{13}C_1$-acetate overnight followed by drinking a fructose/glucose solution the next morning. Blood samples were collected and labeled palmitate was measured in VLDL-TG to evaluate the proportion of fatty acids derived from DNL. TVB-2640 had a significant impact on DNL at all doses tested (Figs. 4 and 5) and was generally well-tolerated in these subjects (Table 2).

RESULTS

Figure 1. Treatment of mice with diet-induced NASH with the FASN inhibitor, TVB-3664*

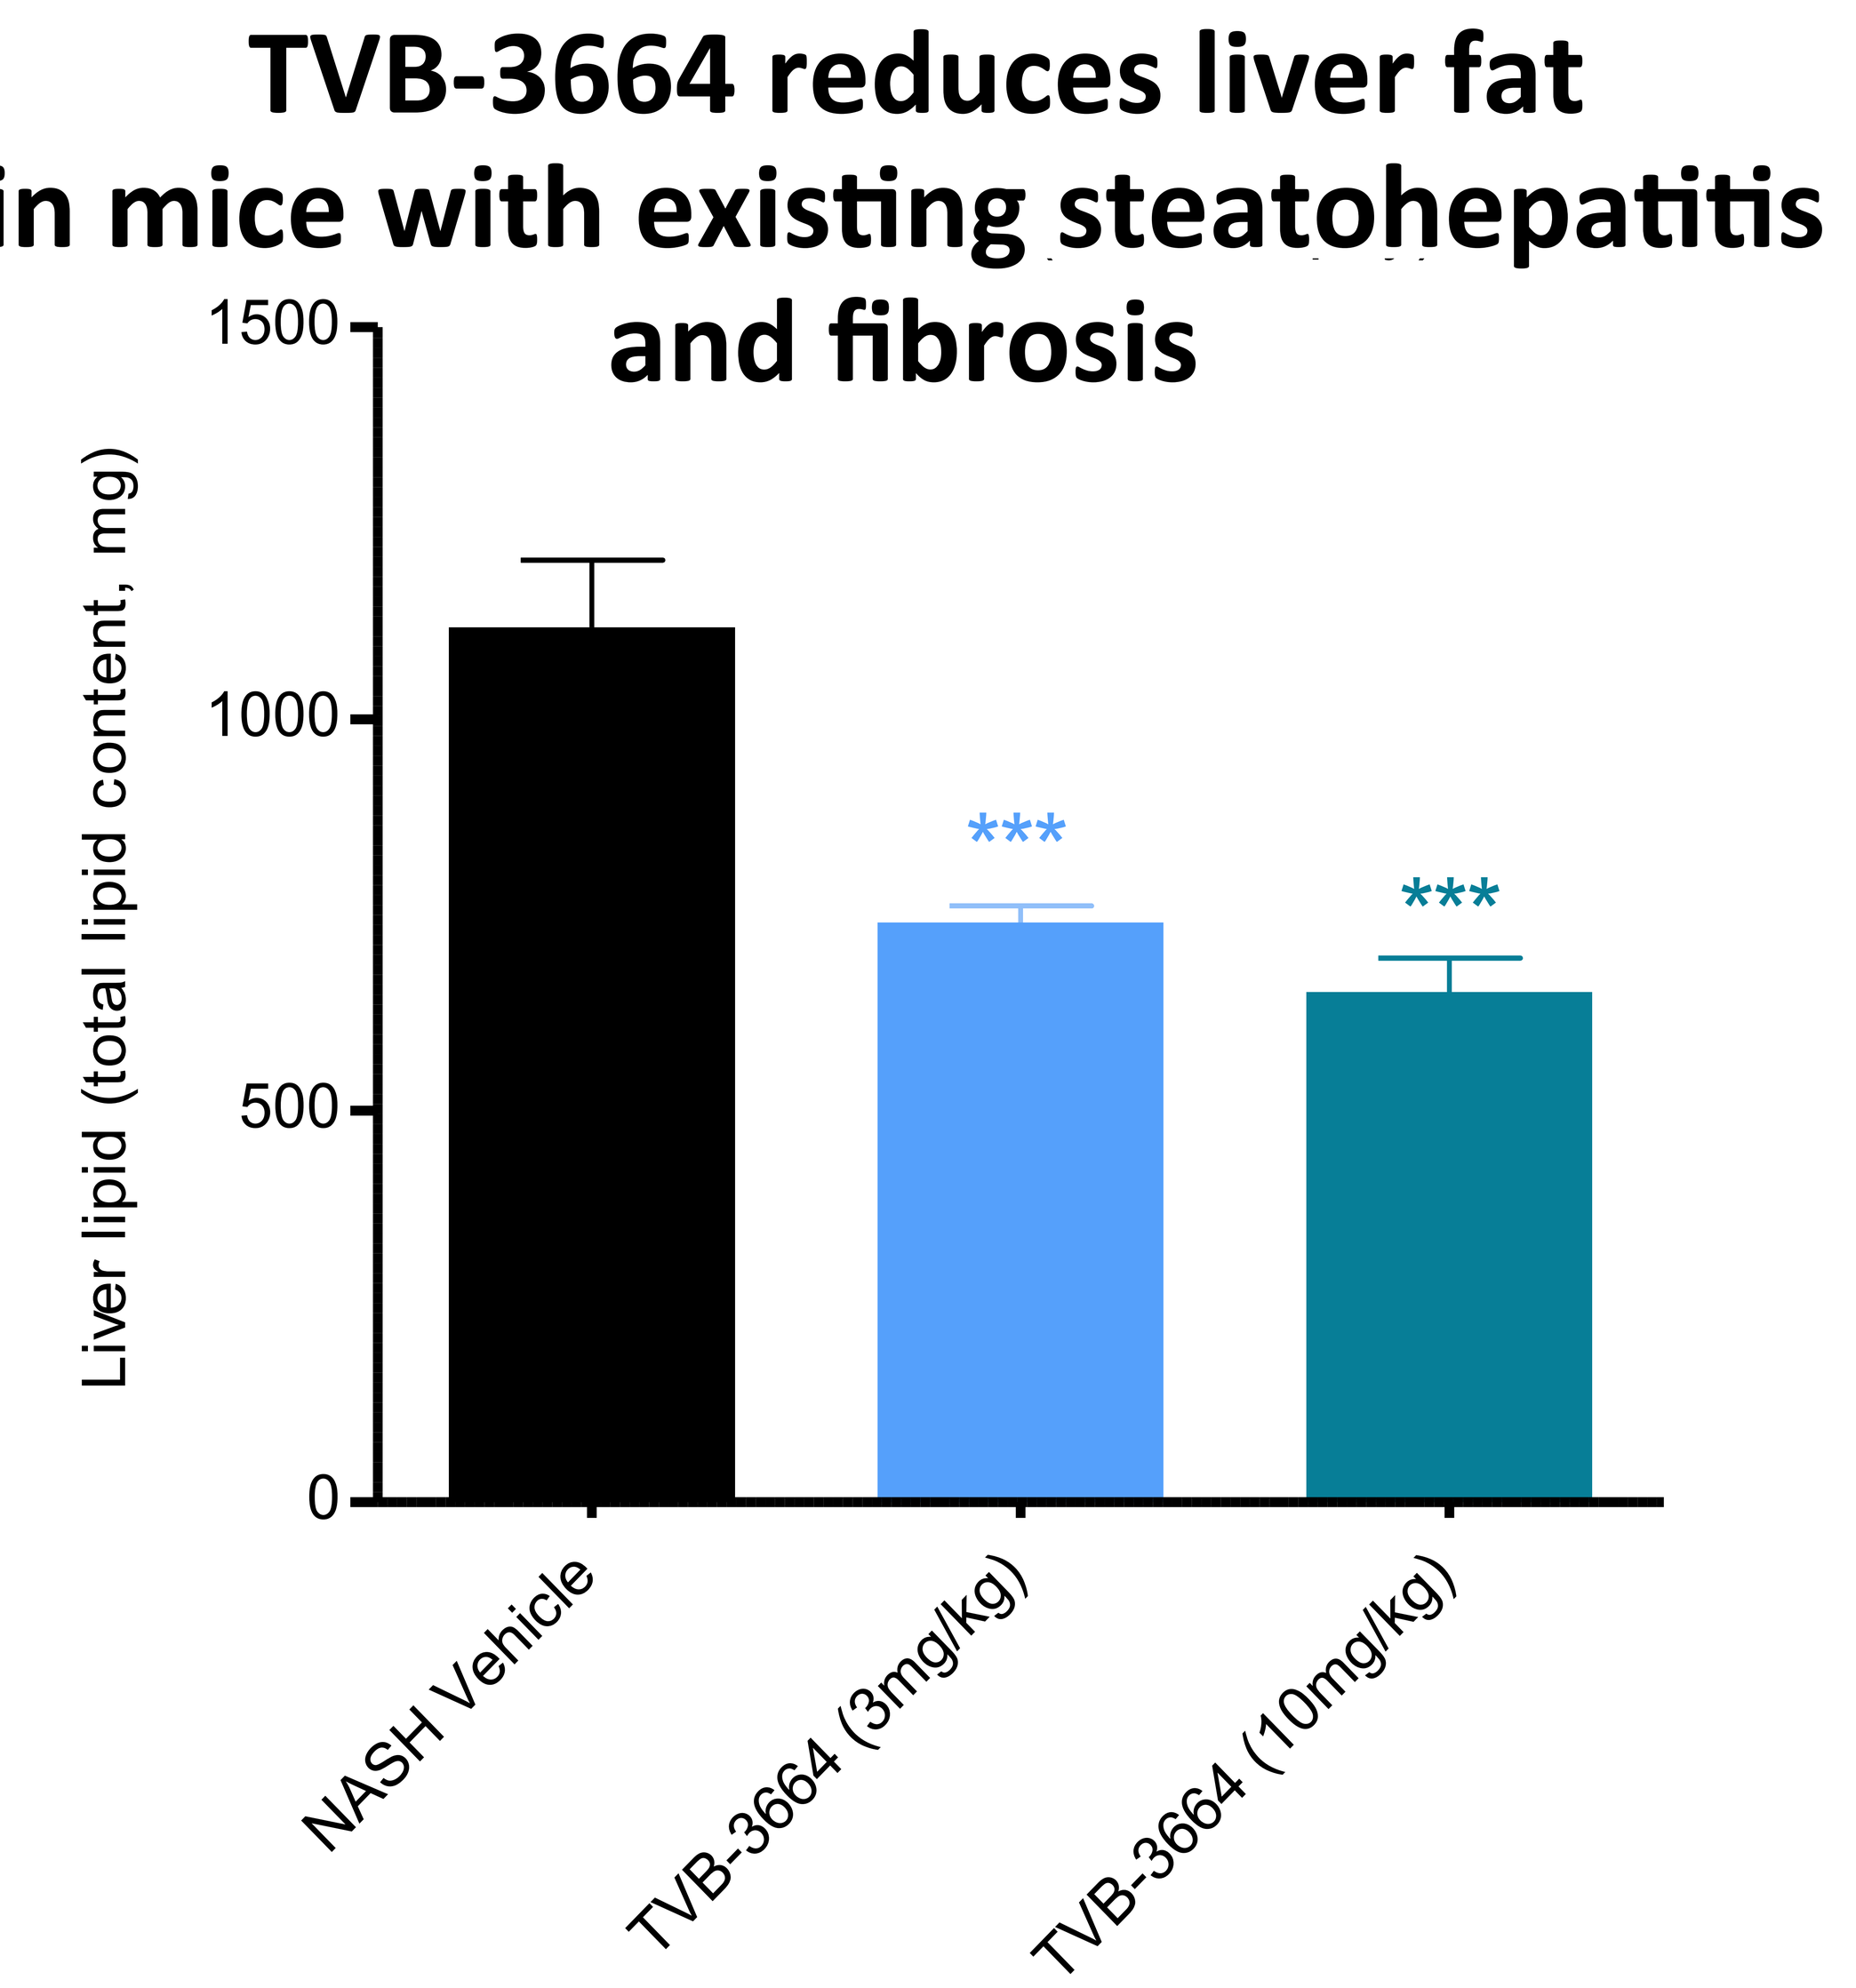
TVB-3664 reduces liver fat in mice with existing steatohepatitis and fibrosis
Liver lipid (total lipid content, mg)
1500
1000
500
0

NASH Vehicle
TVB-3664 (3mg/kg)
TVB-3664 (10mg/kg)

* TVB-3664, an analog of TVB-2640, is used in murine studies due to enhanced PK & potency in mice.

Figure 2. A single oral dose of TVB-2640 (50mg) is well absorbed and has a half-life of >9 hours

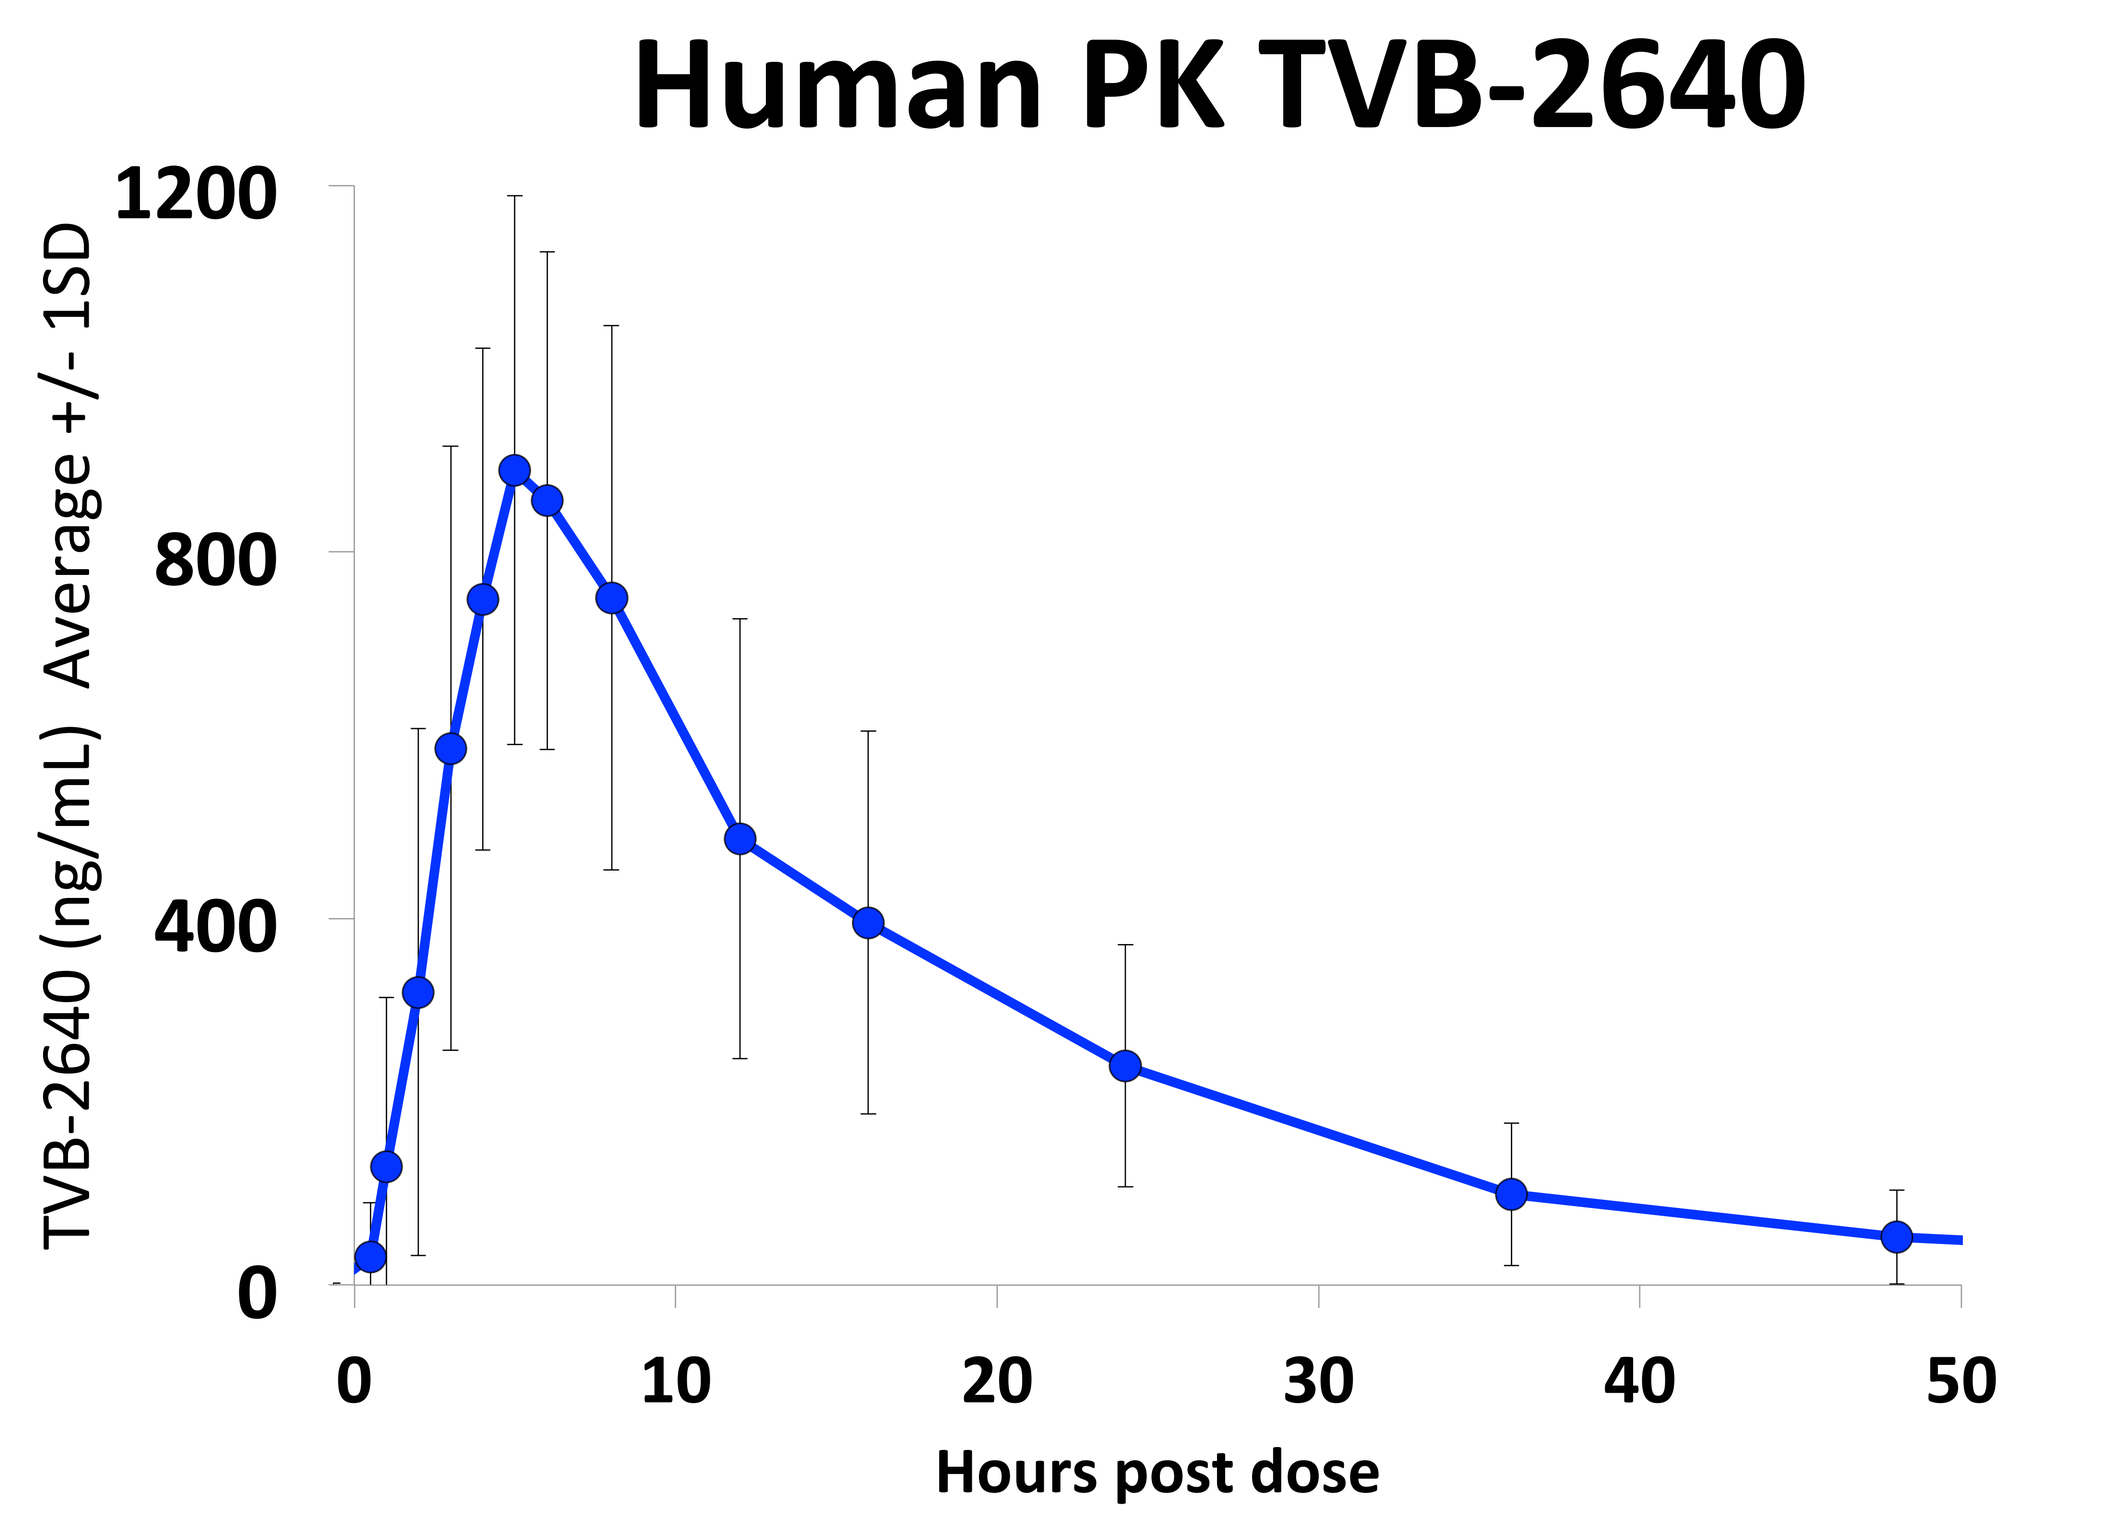
Human PK TVB-2640
TVB-2640 (ng/mL) Average +/- 1SD
1200
800
400
0
0
10
20
30
40
50
Hours post dose

Healthy volunteers (n=20) were dosed with one 50mg capsule of TVB-2640 in the fasted state and blood collected for 48 hours.

Table 1. Demographics of subjects in the clinical pharmacology study (males only)

N	6	2
Dose	50mg	150mg
	Mean (*range*)	Mean (*range*)
BMI	37 (*31-40*)	38 (*34-41*)
Fasting glucose (mg/dL)	100 (*89-115*)	100 (*98-101*)
Triglycerides (mg/dL)	205 (*151-317*)	230 (*155-304*)
Insulin (IU/ml)	16 (*10-30*)	13 (*6-19*)

Figure 3. Pharmacology study outline

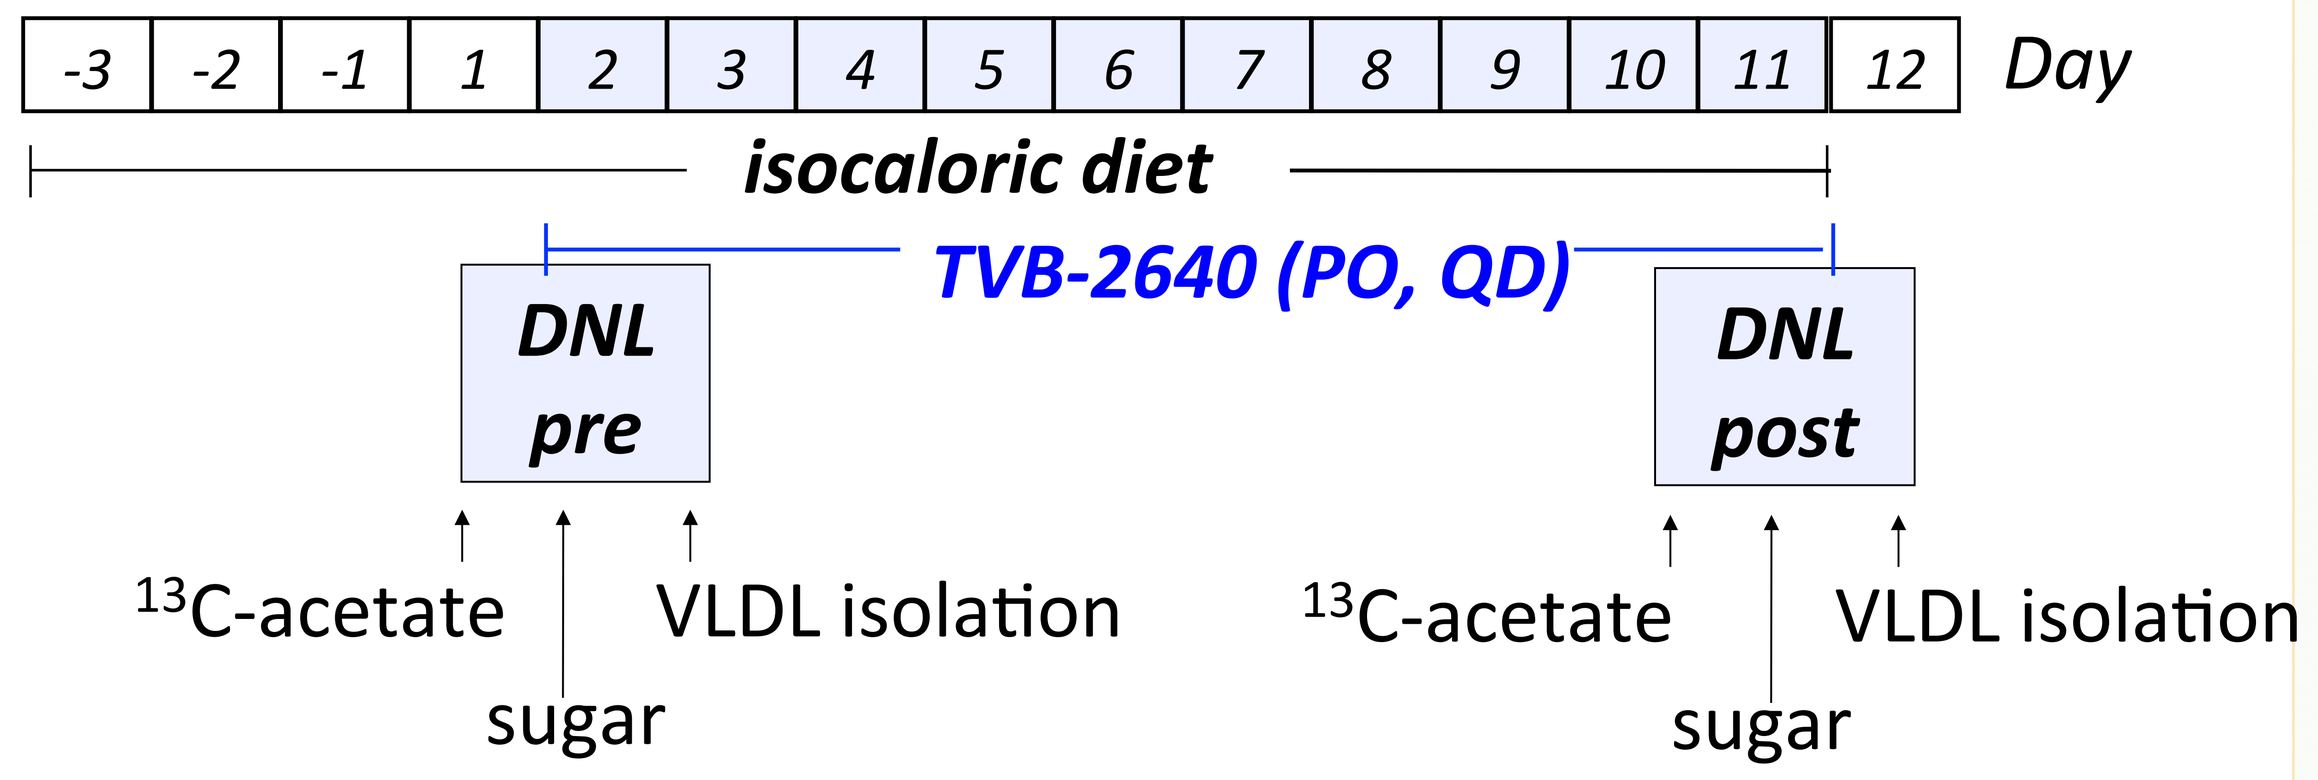
-3 -2 -1 1 2 3 4 5 6 7 8 9 10 11 12 Day
isocaloric diet
TVB-2640 (PO, QD)
DNL pre
DNL post
13C-acetate
sugar
VLDL isolation
13C-acetate
sugar
VLDL isolation

Figure 4. TVB-2640 significantly reduces DNL

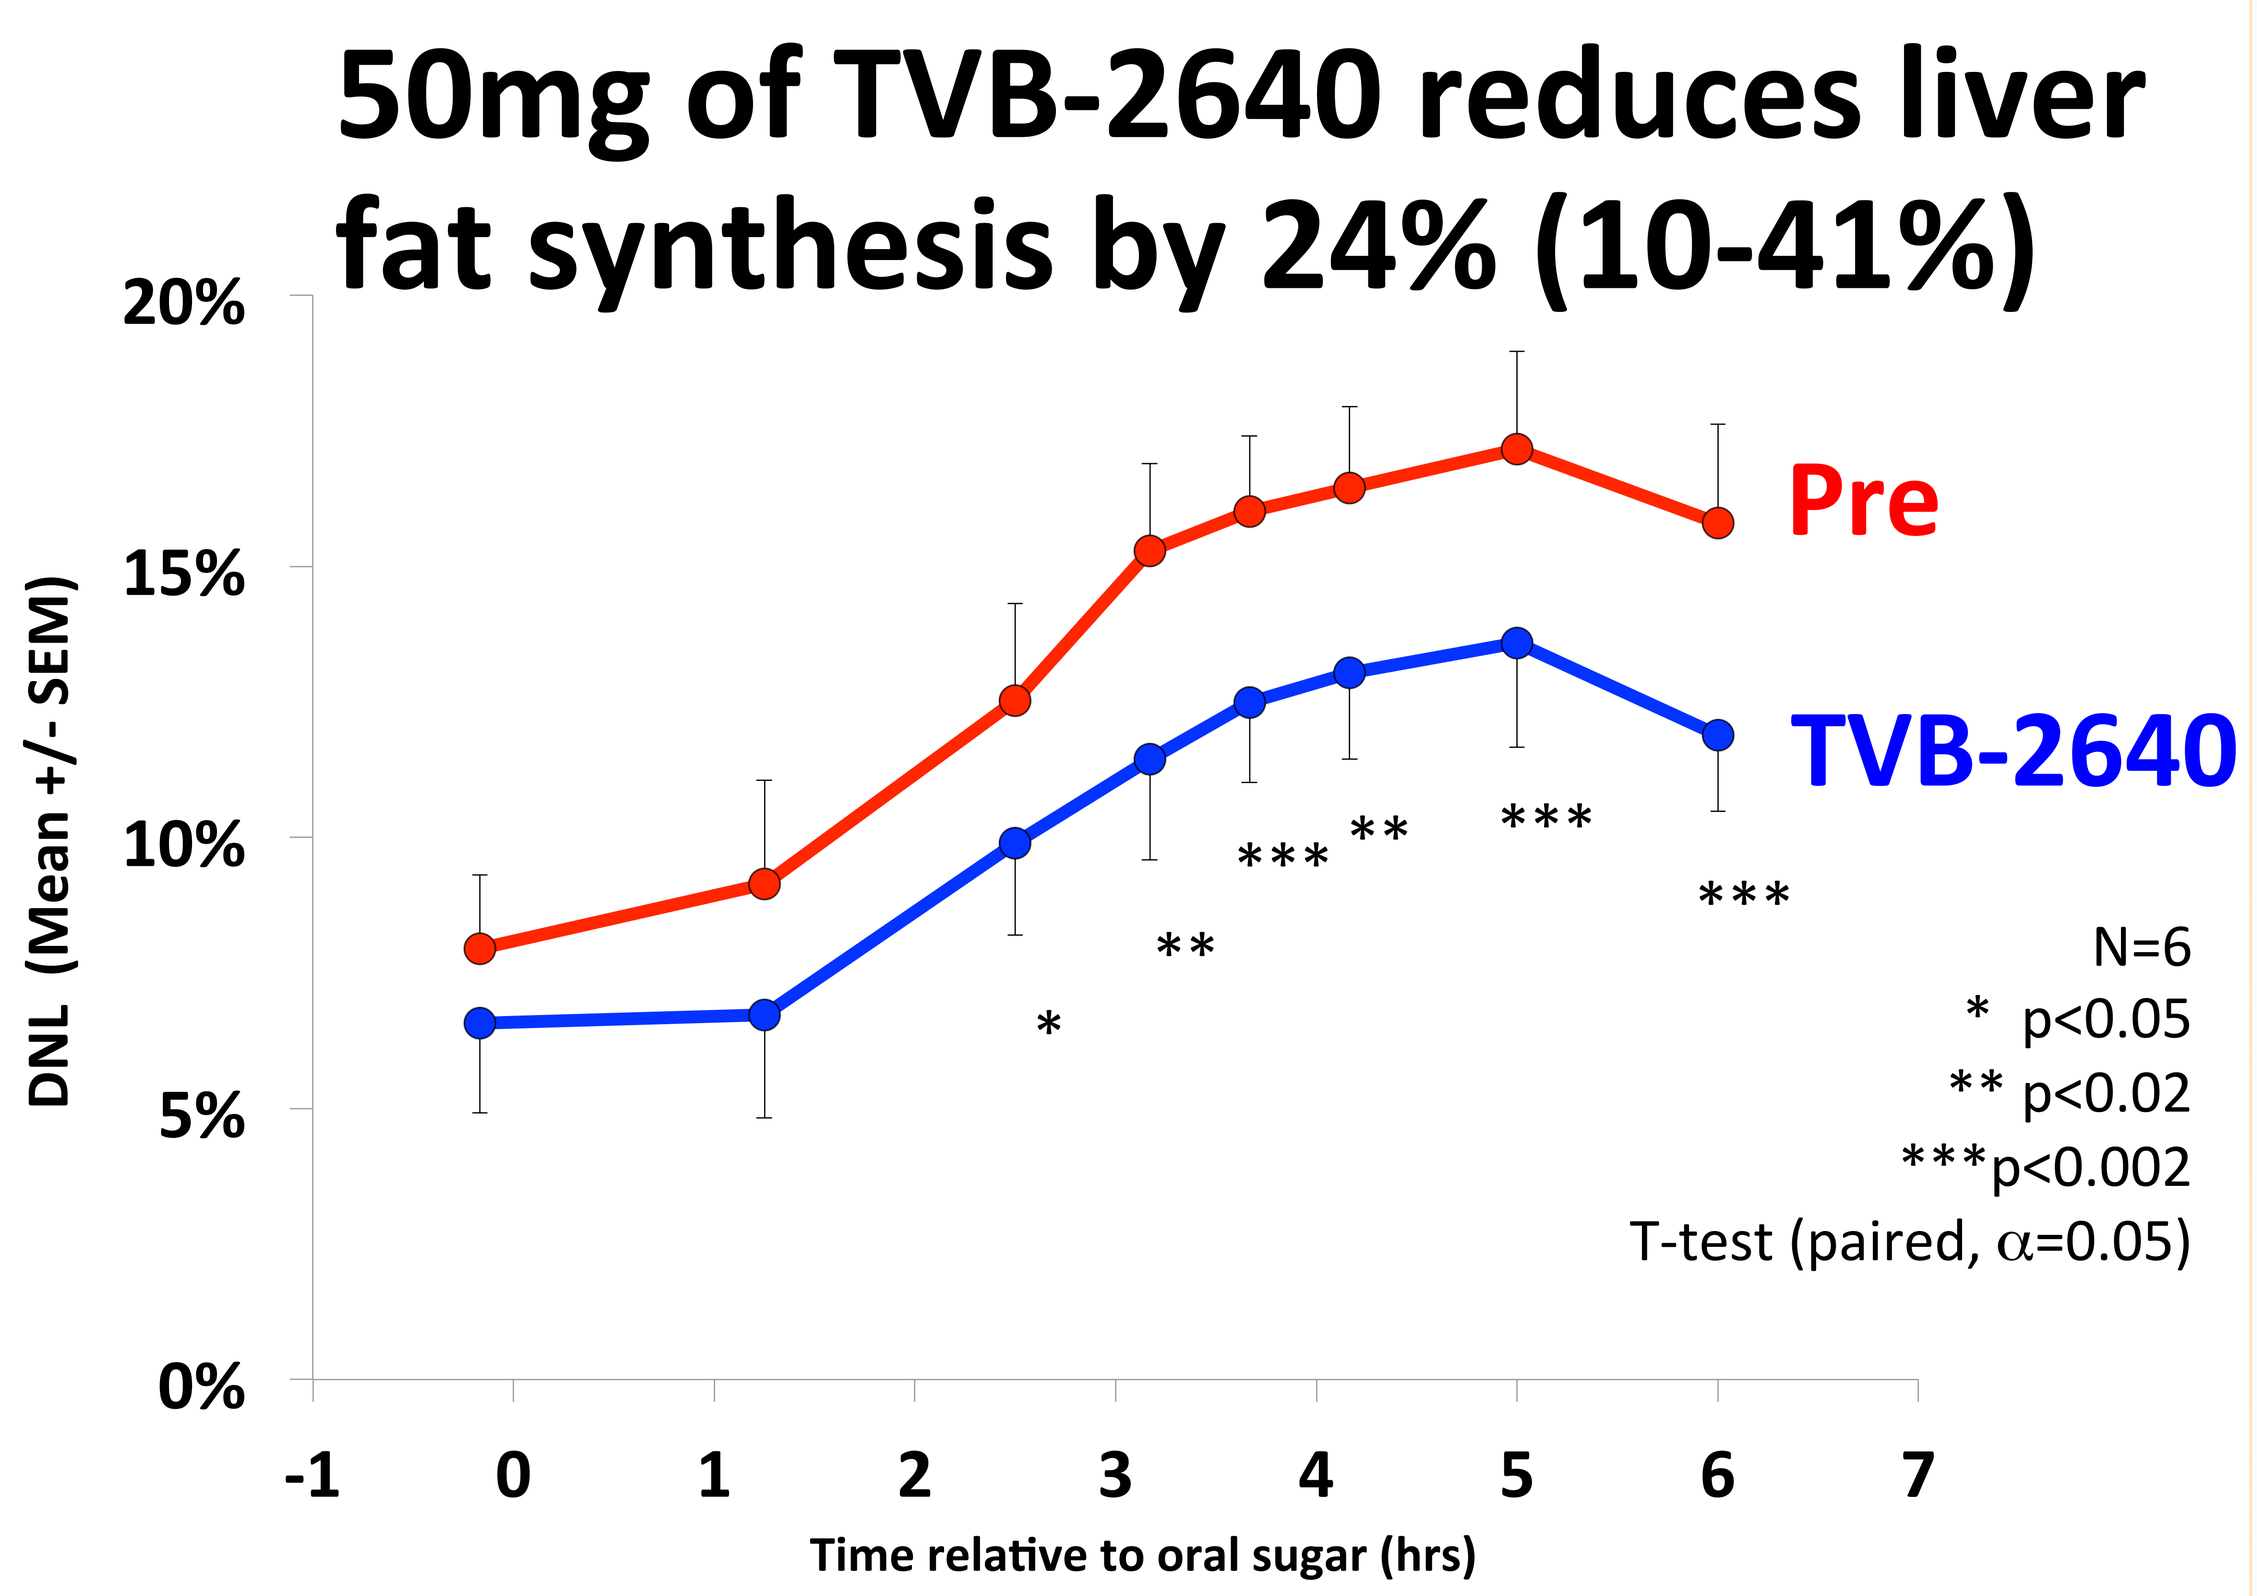
50mg of TVB-2640 reduces liver fat synthesis by 24% (10-41%)
DNL (Mean +/- SEM)
20%
15%
10%
5%
0%
Pre
TVB-2640
N=6
* p<0.05
** p<0.02
***p<0.002
T-test (paired, α=0.05)
-1 0 1 2 3 4 5 6 7
Time relative to oral sugar (hrs)

Figure 5. Excellent correlation between drug levels in plasma and reduction of DNL

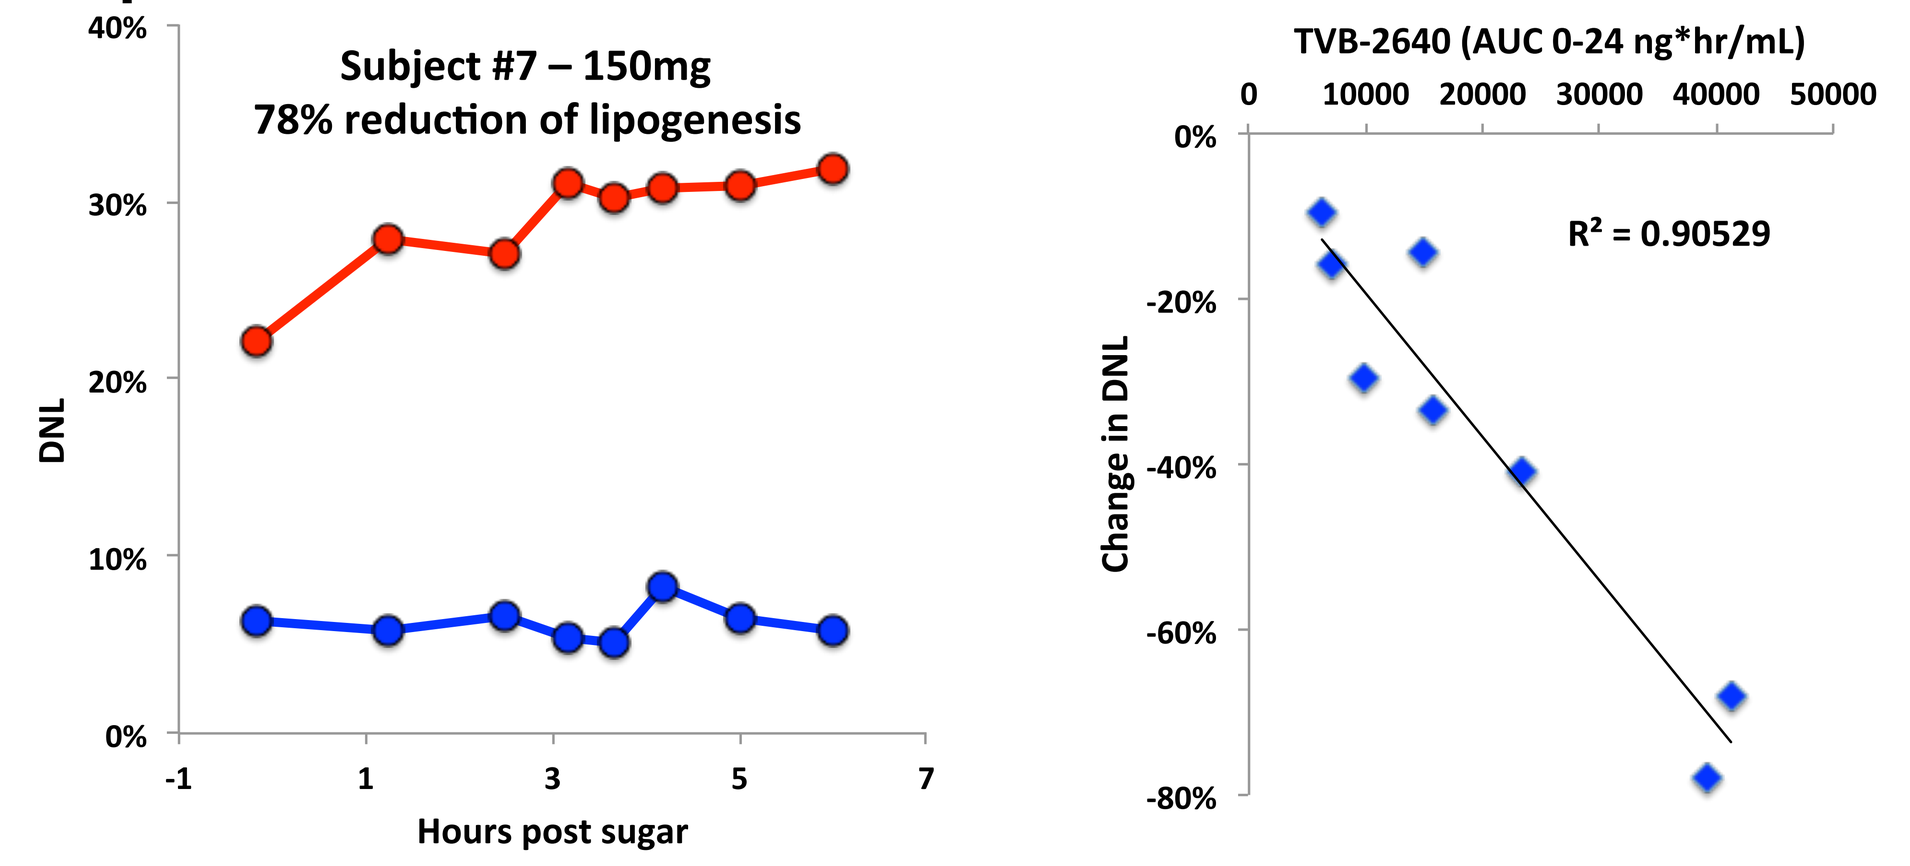
Subject #7 – 150mg
78% reduction of lipogenesis
DNL
Hours post sugar
TVB-2640 (AUC 0-24 ng*hr/mL)
0 10000 20000 30000 40000 50000
Change in DNL
R² = 0.90529

Table 2. TVB-2640 was well tolerated

N	6		2	
Dose	50mg		150mg	
	Pre-value Mean (range)	%Change Mean (range)	Pre-value Mean (range)	%Change Mean (range)
AST[1]	23 (16-8)	-5% (-37-50)	26 (26)	-6%(-31-19)
ALT[1]	32 (17-47)	-13% (-39-21)	36 (35-37)	1%(-35-37)
Triglycerides, clinical chemistry panel [1,2]		No sig change		No sig change
Clinical side-effects	--	None	--	1 of 2 hair thinning

[1] In a study of cancer patients – 54 patients receiving doses of 100-250 daily for 6 – 24 weeks had no significant changes in LFTs, triglycerides, or LDL-cholesterol

[2] Clinical chemistry panel included: glucose, BUN, creatinine, sodium, potassium, chloride, carbon dioxide, calcium, total protein, albumin, alkaline phosphatase, & total bilirubin

CONCLUSIONS

- Pharmacological inhibition of FASN treats NASH in mice fed a high-fat, high-sugar diet.
- Once-daily dosing of TVB-2640, an oral, highly-selective FASN inhibitor, was supported by its PK profile.
- 50mg of TVB-2640 significantly reduced hepatic DNL by an average of 24%.
- 150mg of TVB-2640 in a subject with a high baseline (pre sugar) rate of lipogenesis potently inhibited hepatic DNL.
- Plasma levels of TVB-2640 strongly correlate with the level of suppression of lipogenesis, enabling dose titration.

DISCLOSURES

These studies were sponsored by 3-V Biosciences, Inc.

Contact information George Kemble (George.Kemble@3VBIO.com) or Dennis Hom (Dennis.Hom@3VBIO.com)

AASLD AMERICAN ASSOCIATION FOR THE STUDY OF LIVER DISEASES