Fatty acid synthase (FASN) inhibitor reduces atherosclerosis development in diet-induced dyslipidaemia LDL receptor knockout mice with MASH

Wen-Wei Tsai[1], Eveline Gart[2], Martine C. Morrison[2], Eduardo Martins[1], George Kemble[1], Marie O'Farrell[1]

1. Sagimet Biosciences, San Mateo, United States, 2. TNO Metabolic Health Research, Leiden, Netherlands

Introduction

- Denifanstat (TVB-2640) is an oral, once daily, selective FASN inhibitor in clinical development for MASH
- FASN inhibition targets 3 hallmarks of MASH: inhibits liver fat synthesis & accumulation (hepatocytes), inhibits fibrosis (hepatic stellate cells require DNL for activation) and decreases inflammation (inflammasome activation by palmitate)[1]
- Denifanstat has recently demonstrated statistically significant MASH resolution and fibrosis improvement in the Phase 2b MASH study, FASCINATE-2 (NCT04906421)[2]
- Denifanstat decreased LDL-cholesterol and increased polyunsaturated triglycerides in both Phase 2a FASCINATE-1[3] and Phase 2b FASCINATE-2[2] studies, suggesting potential cardiovascular benefits in addition to direct liver effects

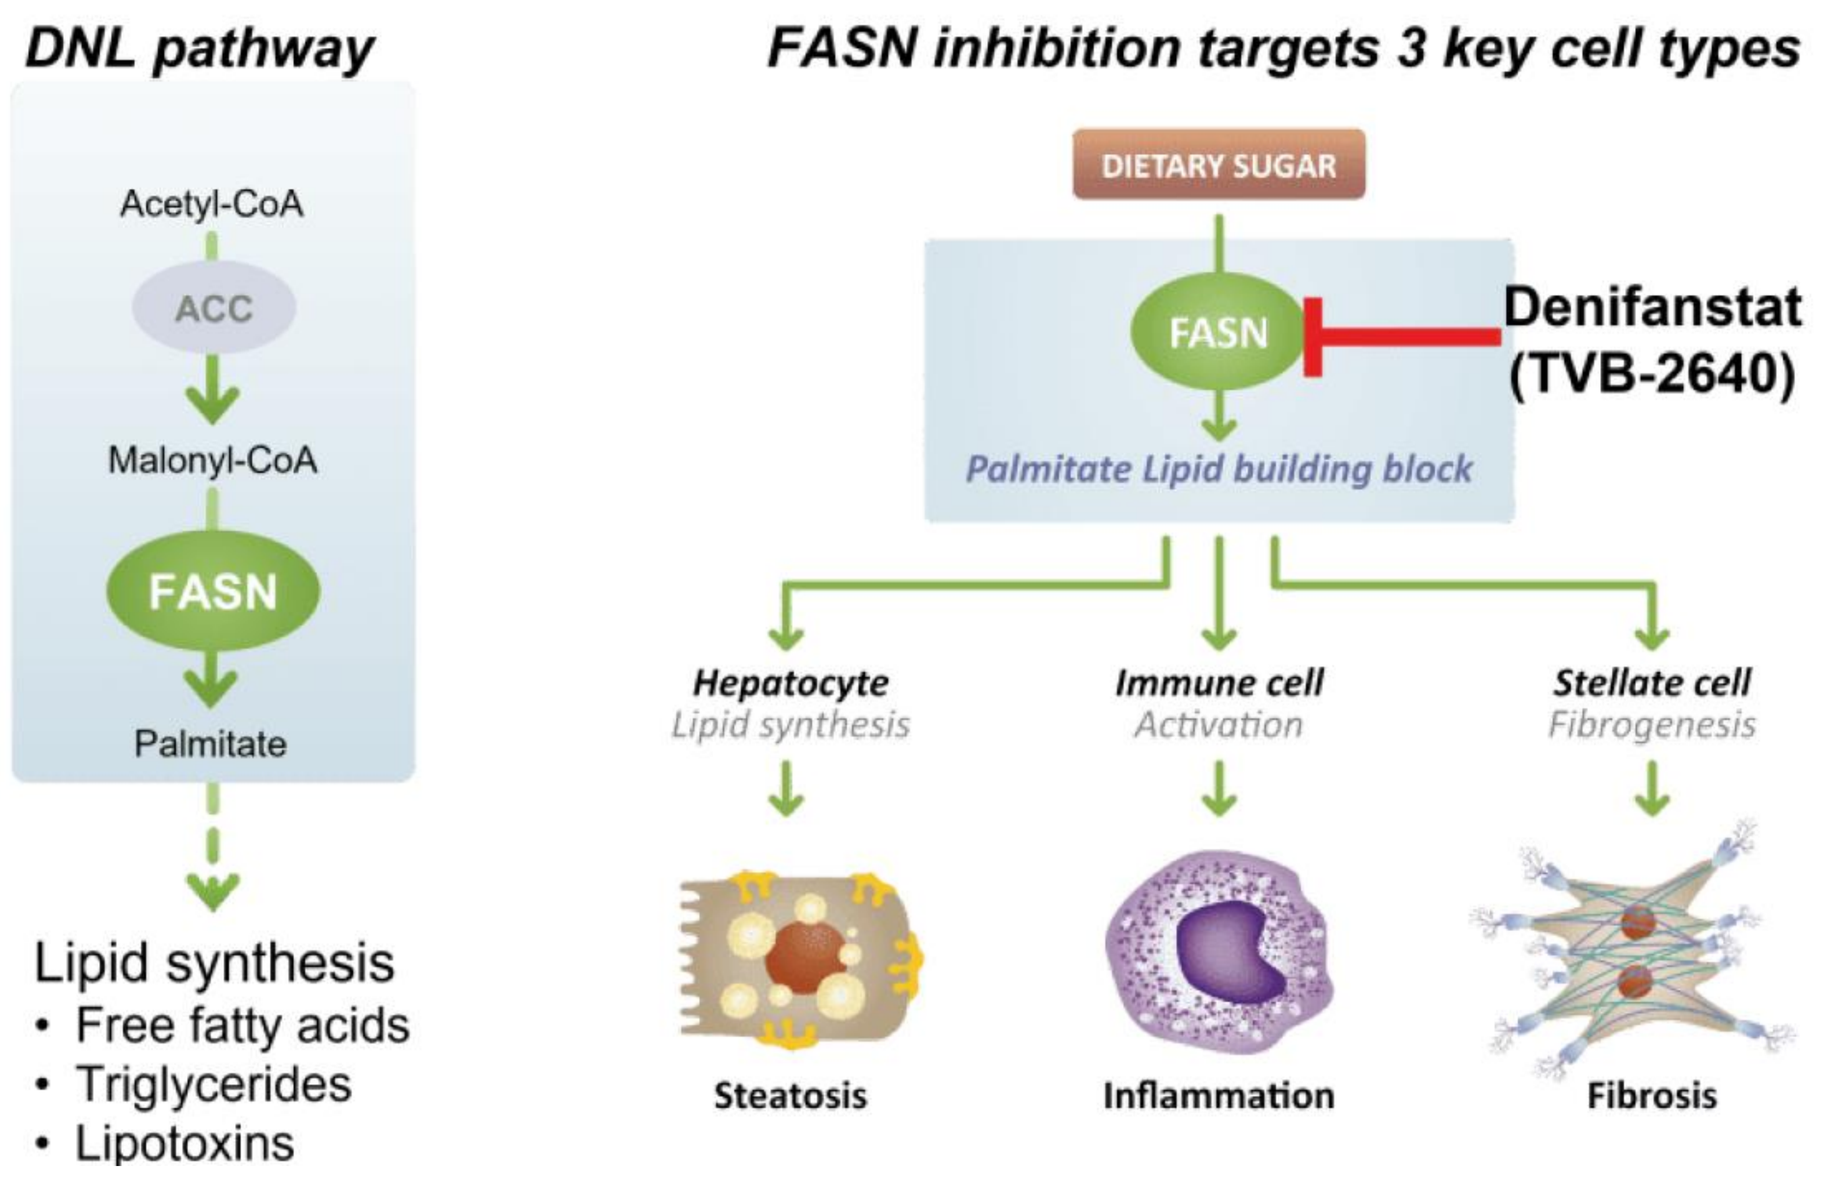
DNL pathway
Acetyl-CoA
ACC
Malonyl-CoA
FASN
Palmitate
Lipid synthesis
• Free fatty acids
• Triglycerides
• Lipotoxins
FASN inhibition targets 3 key cell types
DIETARY SUGAR
FASN
Denifanstat (TVB-2640)
Palmitate Lipid building block
Hepatocyte
Lipid synthesis
Immune cell
Activation
Stellate cell
Fibrogenesis
Steatosis
Inflammation
Fibrosis

Hypothesis & Aims

- Hypothesis: FASN inhibitor would prevent atherosclerosis through reduction of circulating cholesterol in the context of MASH
- This study was to evaluate the effect of FASN inhibitor on plasma lipids and atherosclerosis in LDL receptor knockout (Ldlr-/-) mice with dyslipidaemia and MASH
- The effect of FASN inhibitor on circulating inflammatory factors and liver histology was also evaluated

Methods

- Male Ldlr-/-.Leiden mice were fed with fast-food diet (FFD) for 18 weeks to induce dyslipidaemia, atherosclerosis and MASH features, and treated with TVB-3664 (a surrogate FASN inhibitor for denifanstat, 5 mg/kg, PO, QD) for 10 weeks. Endpoints included plasma lipids, lipoprotein profile, inflammatory marker profile, liver histology and histological analysis of atherosclerosis in the aortic root (lesion area and severity according to the AHA score in 4 cross sections, *TNO, Netherlands*).

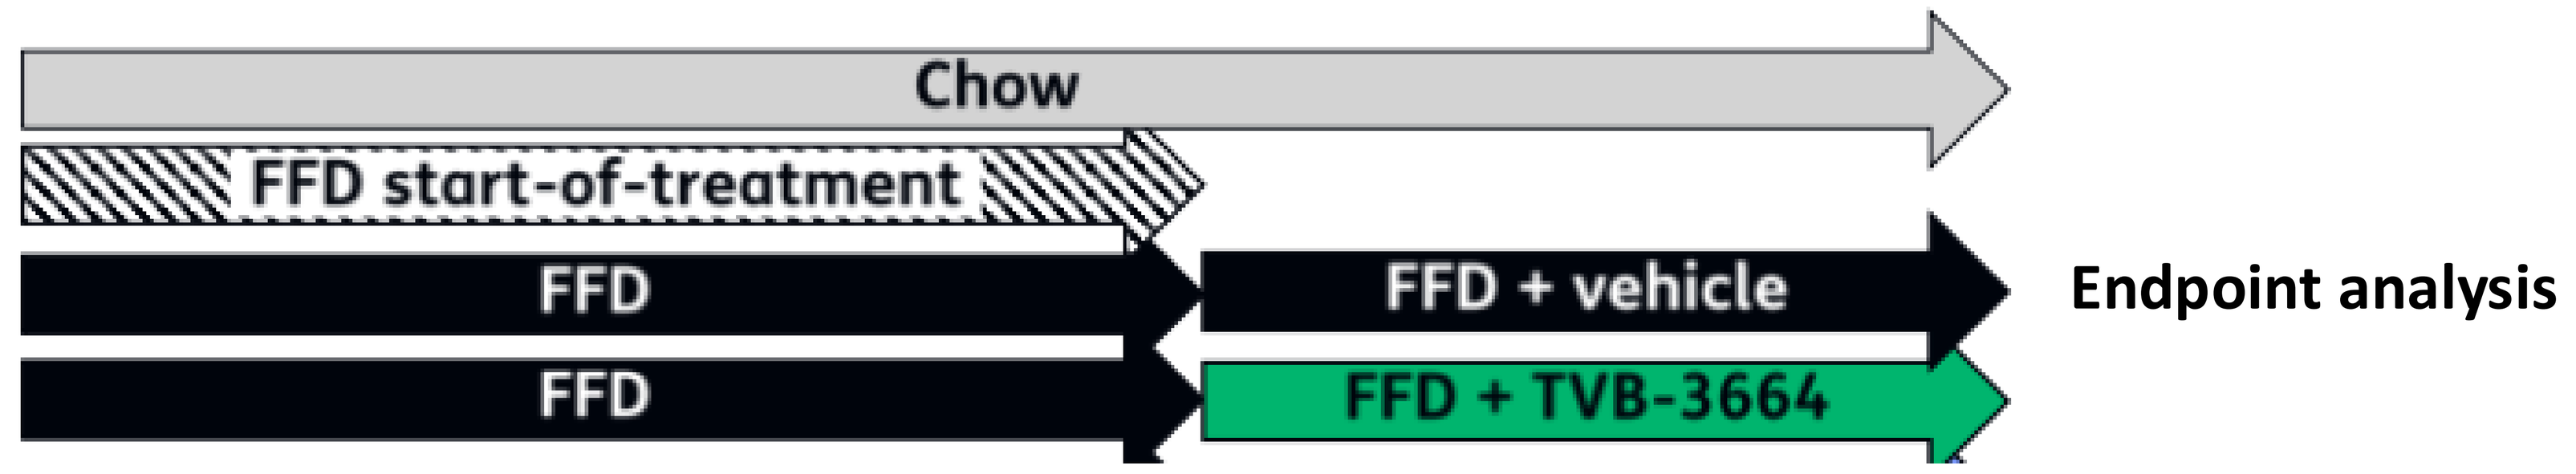
Chow
FFD start-of-treatment
FFD
FFD + vehicle
FFD
FFD + TVB-3664
Endpoint analysis

Results

FASN inhibitor rapidly reduced circulating cholesterol and triglycerides in Ldlr-/- MASH mice

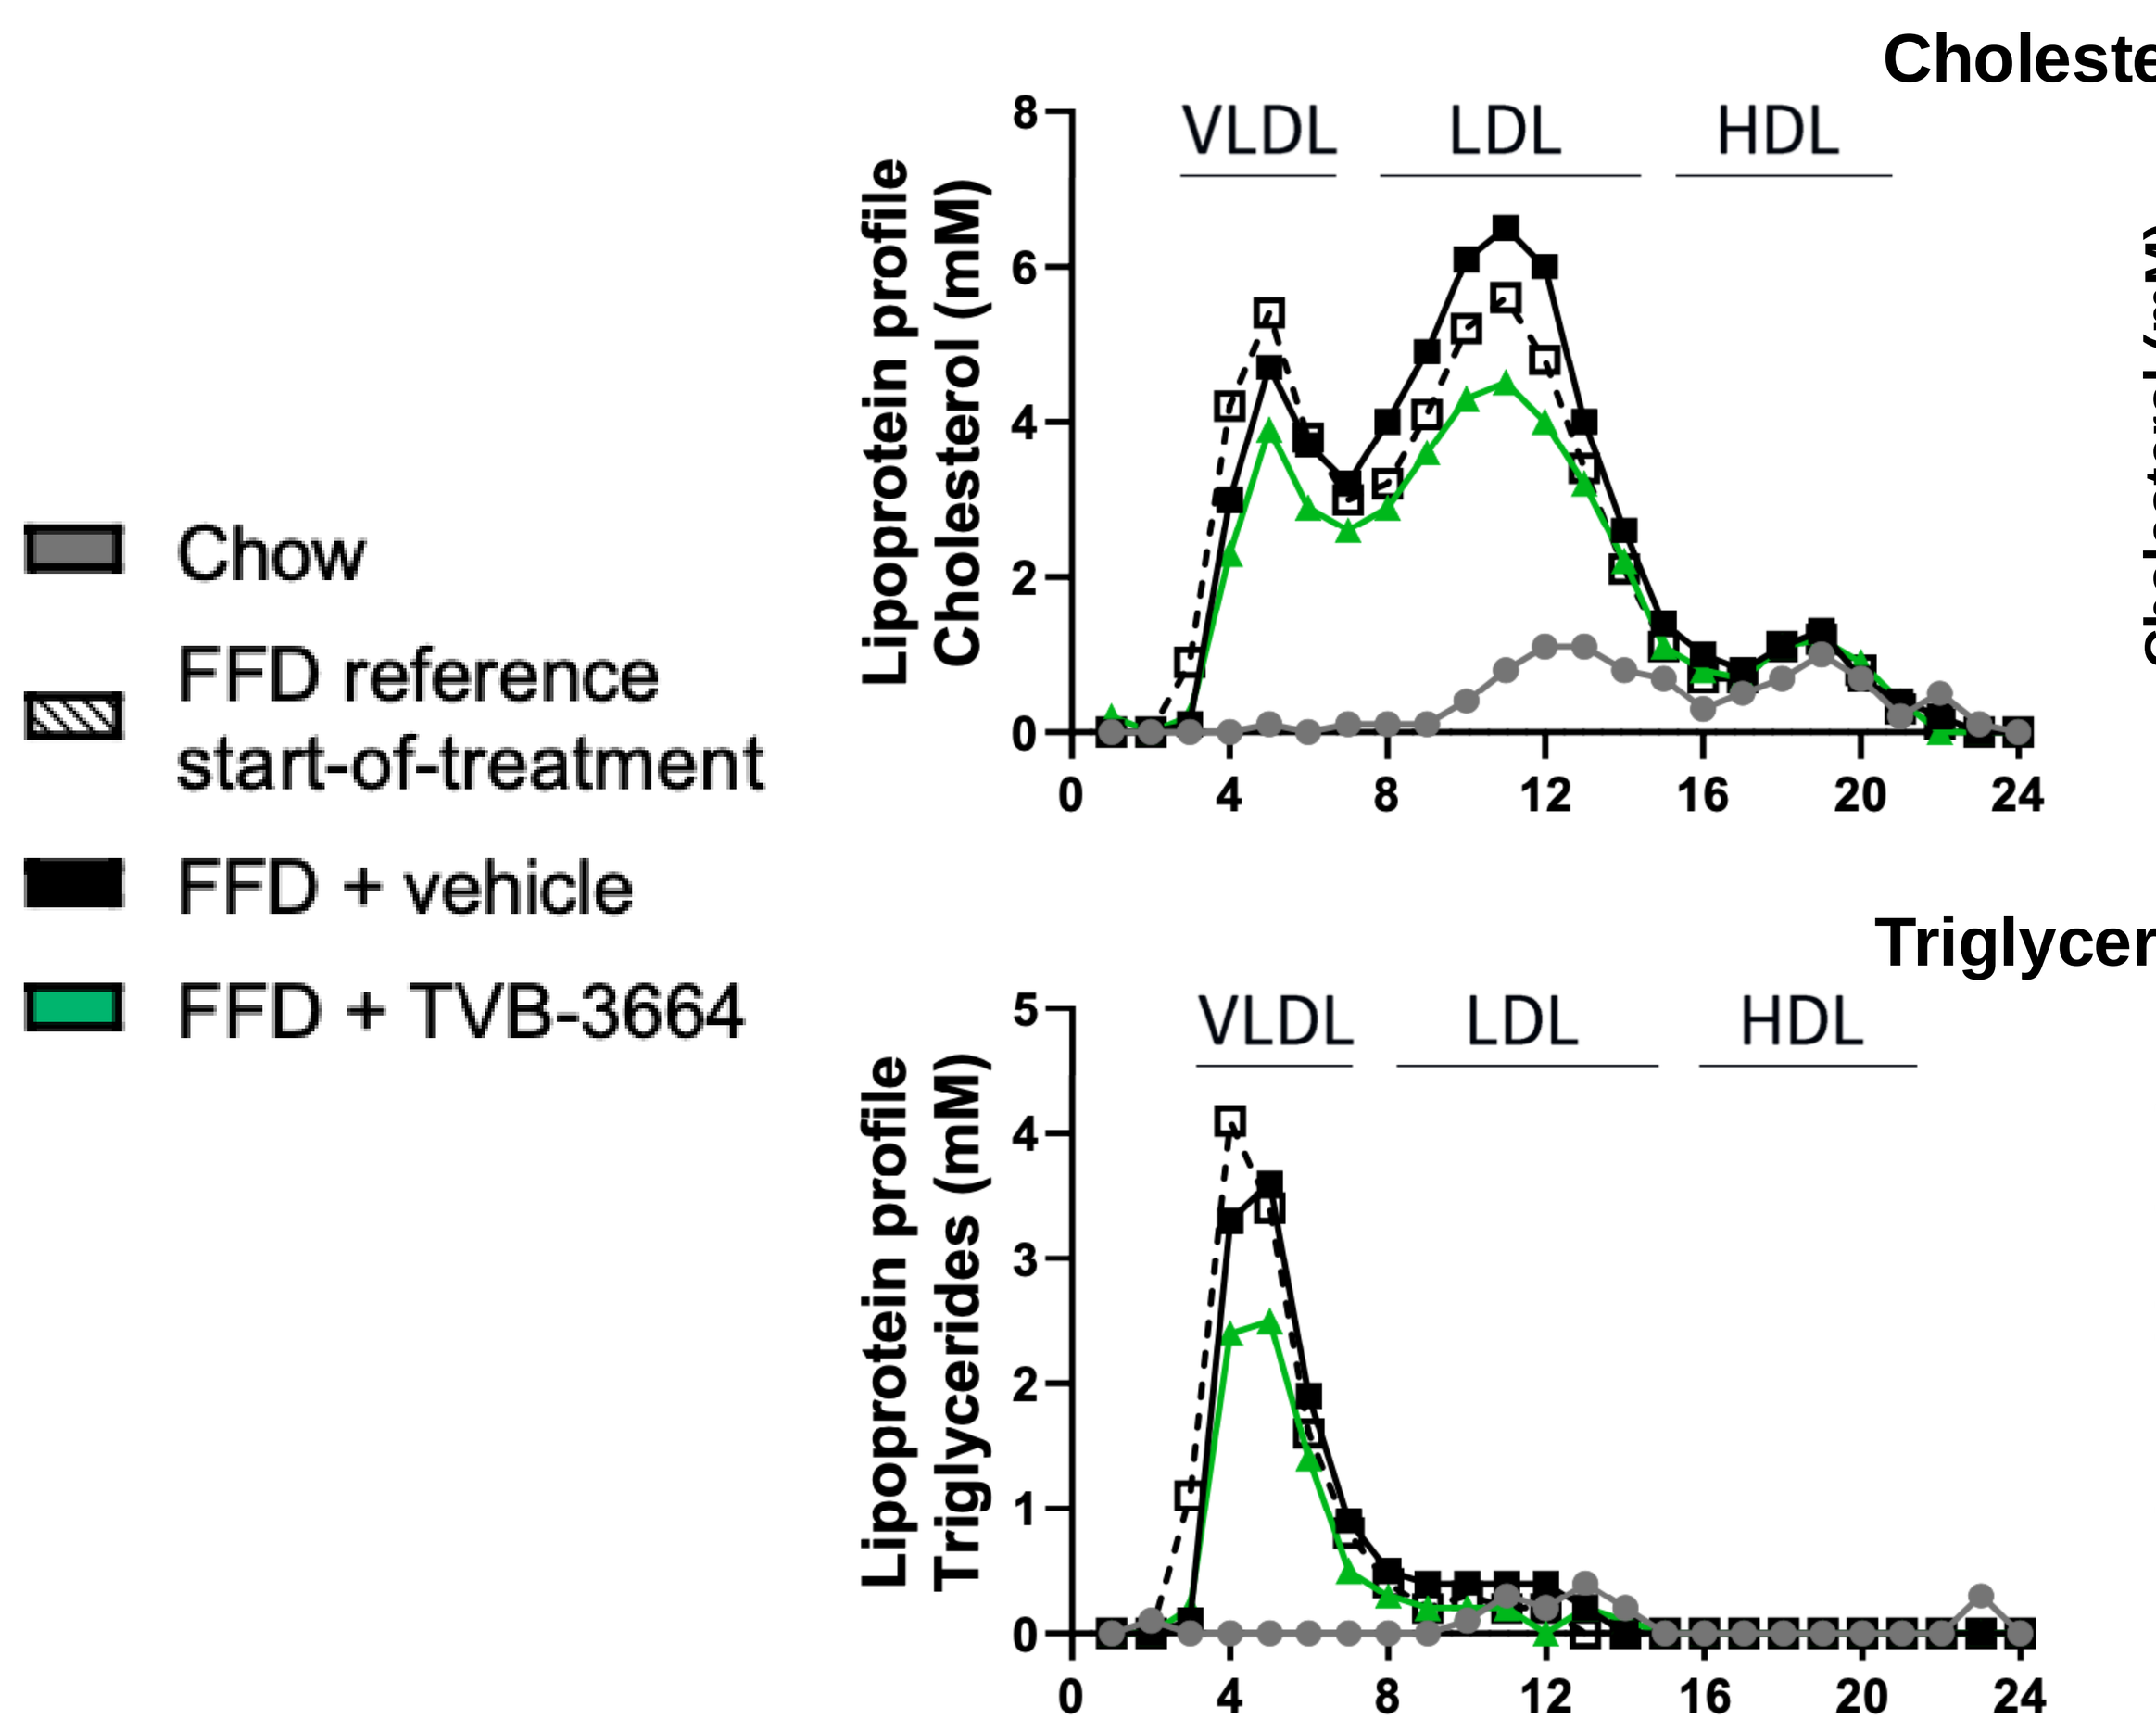
Chow
FFD reference start-of-treatment
FFD + vehicle
FFD + TVB-3664
Cholesterol
Lipoprotein profile Cholesterol (mM)
VLDL
LDL
HDL
Cholesterol (mM)
Triglycerides
Lipoprotein profile Triglycerides (mM)
Triglycerides (mM)

FASN inhibitor reduced several inflammatory markers related to atherosclerosis in Ldlr-/- MASH mice

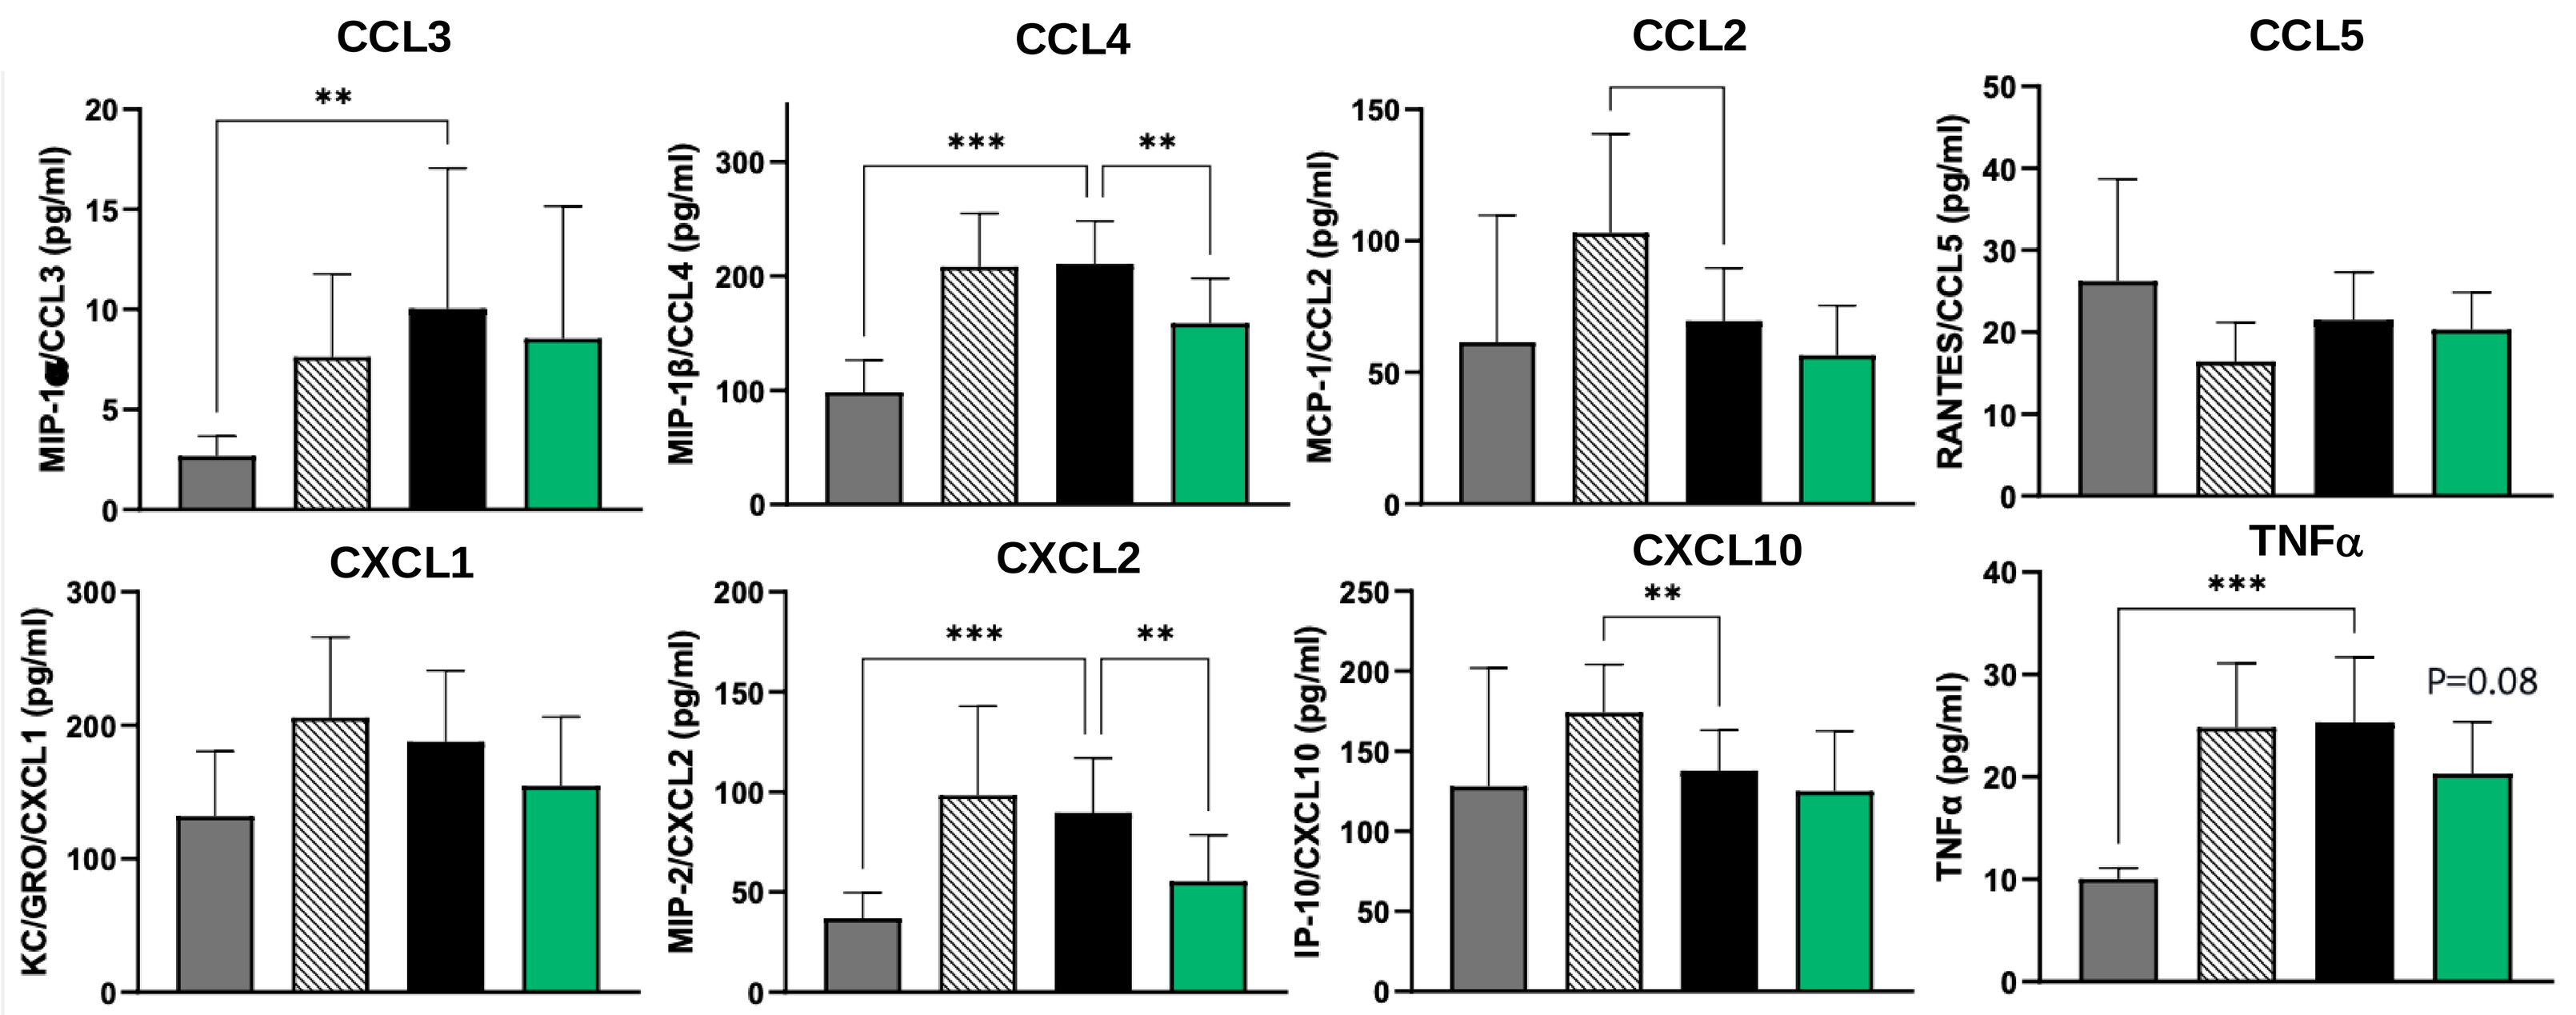
CCL3
MIP-1α/CCL3 (pg/ml)
CCL4
MIP-1β/CCL4 (pg/ml)
CCL2
MCP-1/CCL2 (pg/ml)
CCL5
RANTES/CCL5 (pg/ml)
CXCL1
KC/GRO/CXCL1 (pg/ml)
CXCL2
MIP-2/CXCL2 (pg/ml)
CXCL10
IP-10/CXCL10 (pg/ml)
TNFα
TNFα (pg/ml)
P=0.08

FASN inhibitor reduced total atherosclerotic lesion area per cross-section of aortic root

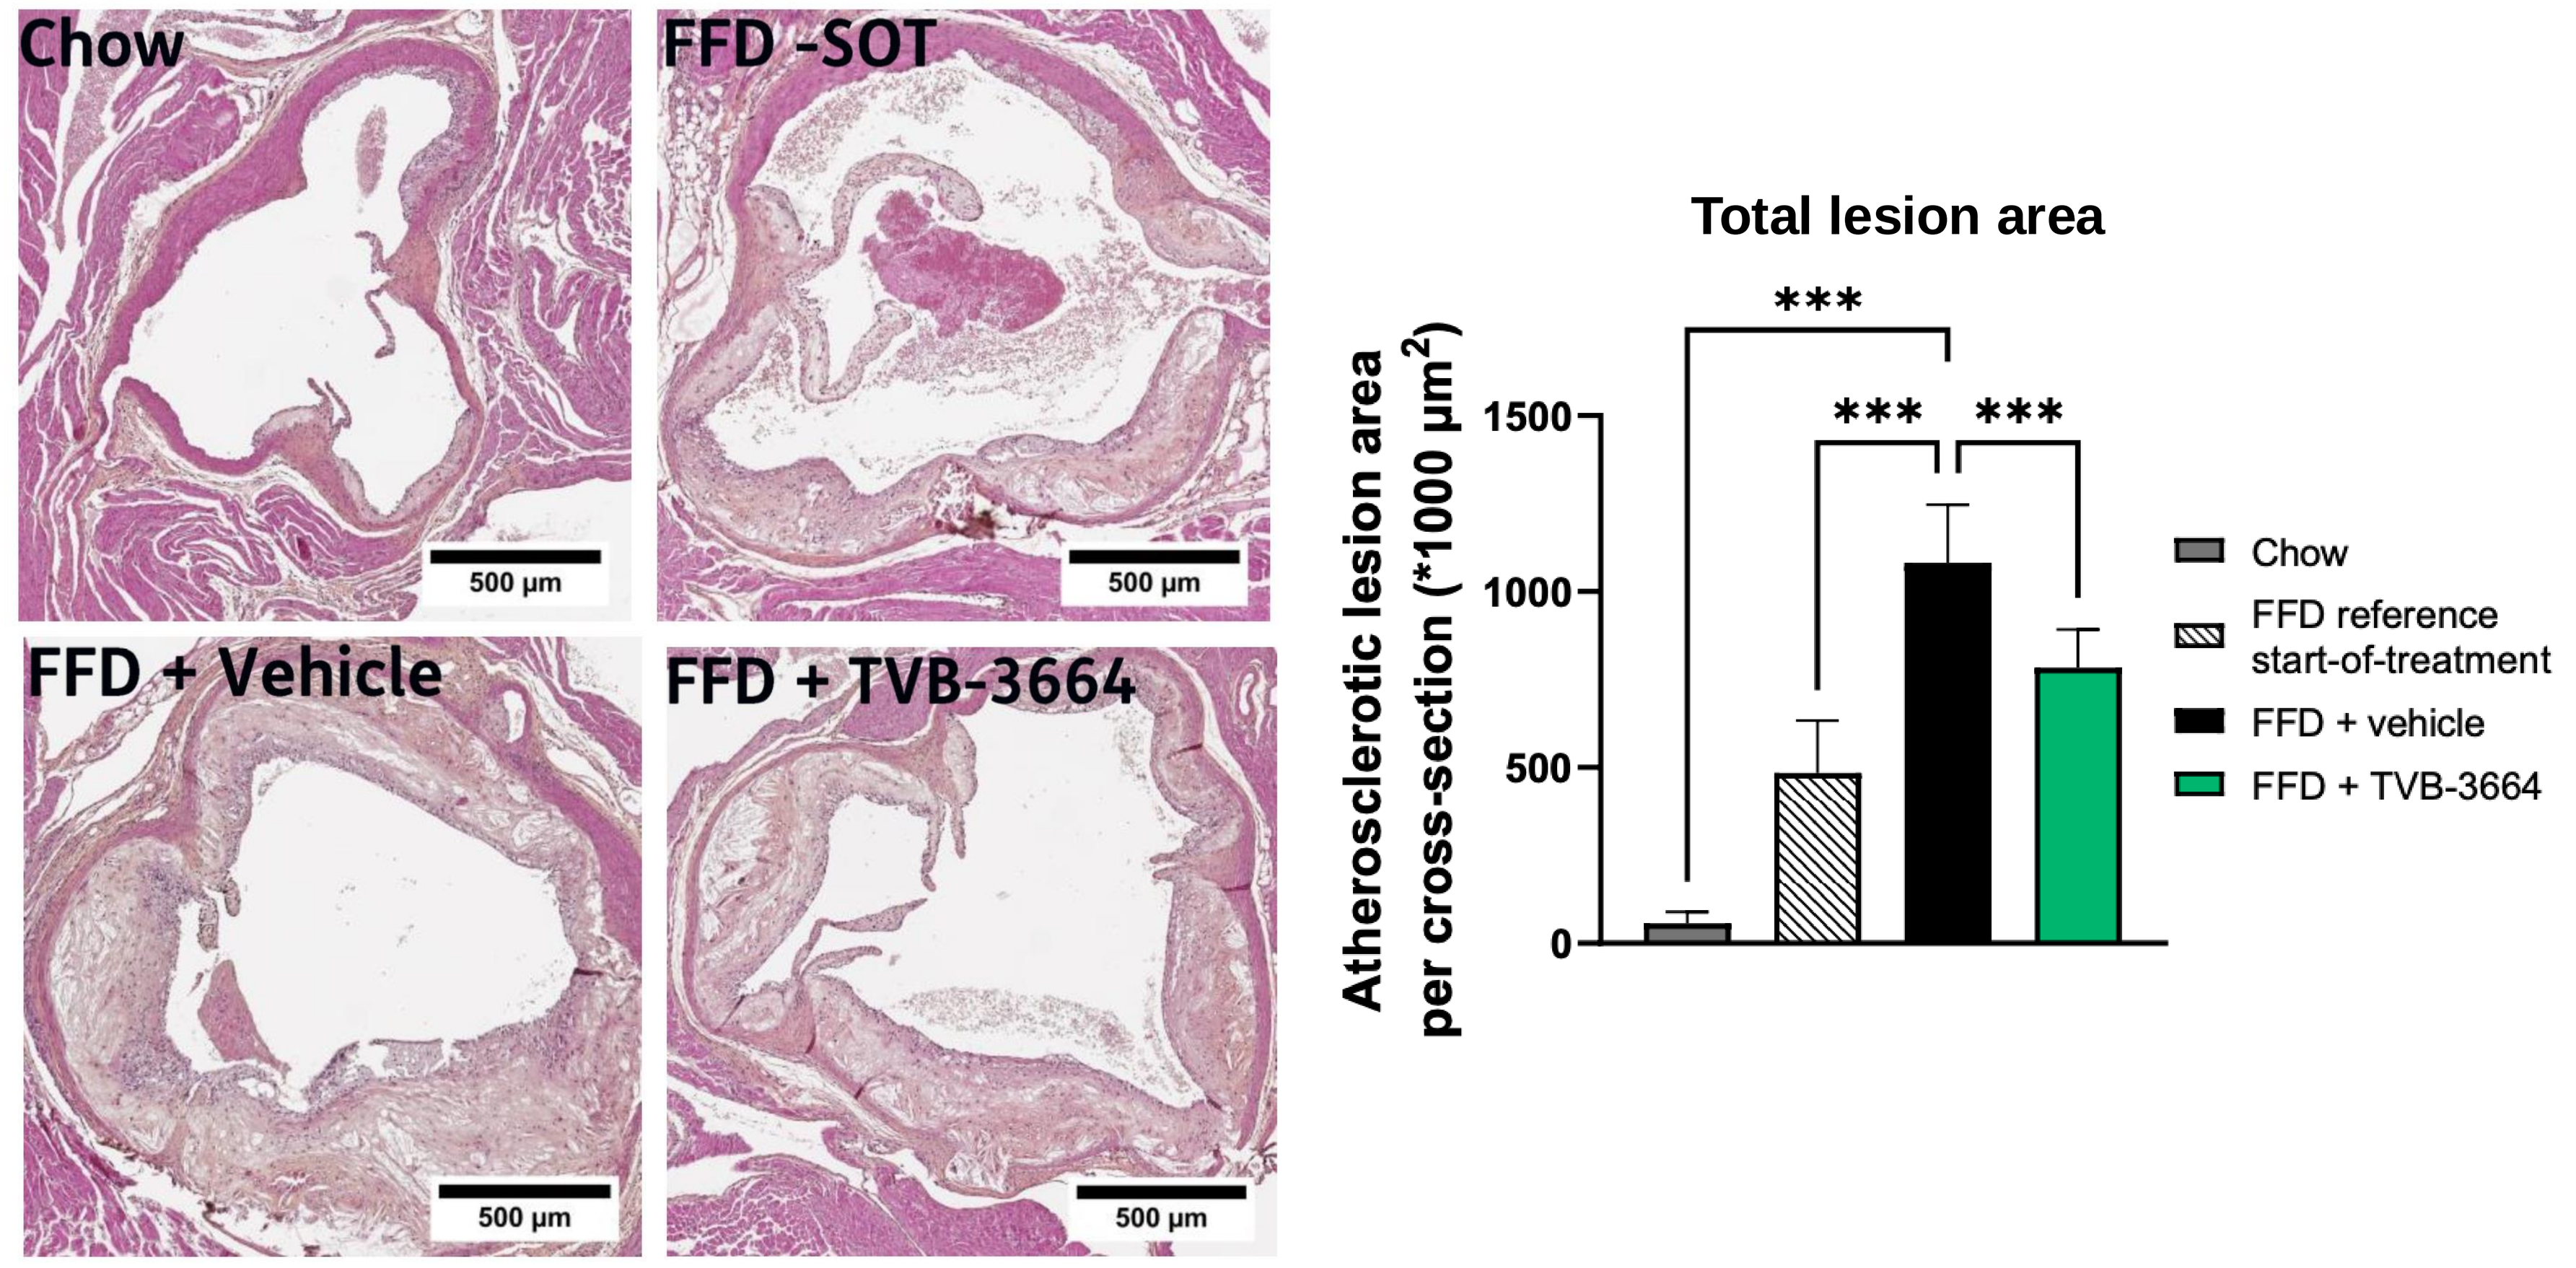
Chow
FFD -SOT
FFD + Vehicle
FFD + TVB-3664
500 µm
Total lesion area
Atherosclerotic lesion area per cross-section (*1000 µm²)
Chow
FFD reference start-of-treatment
FFD + vehicle
FFD + TVB-3664

The Ldlr-/- mice developed severe type V plaque lesions and increased atherosclerotic lesion area with 28-week FFD feeding

FASN inhibitor improved liver histology in Ldlr-/- MASH mice

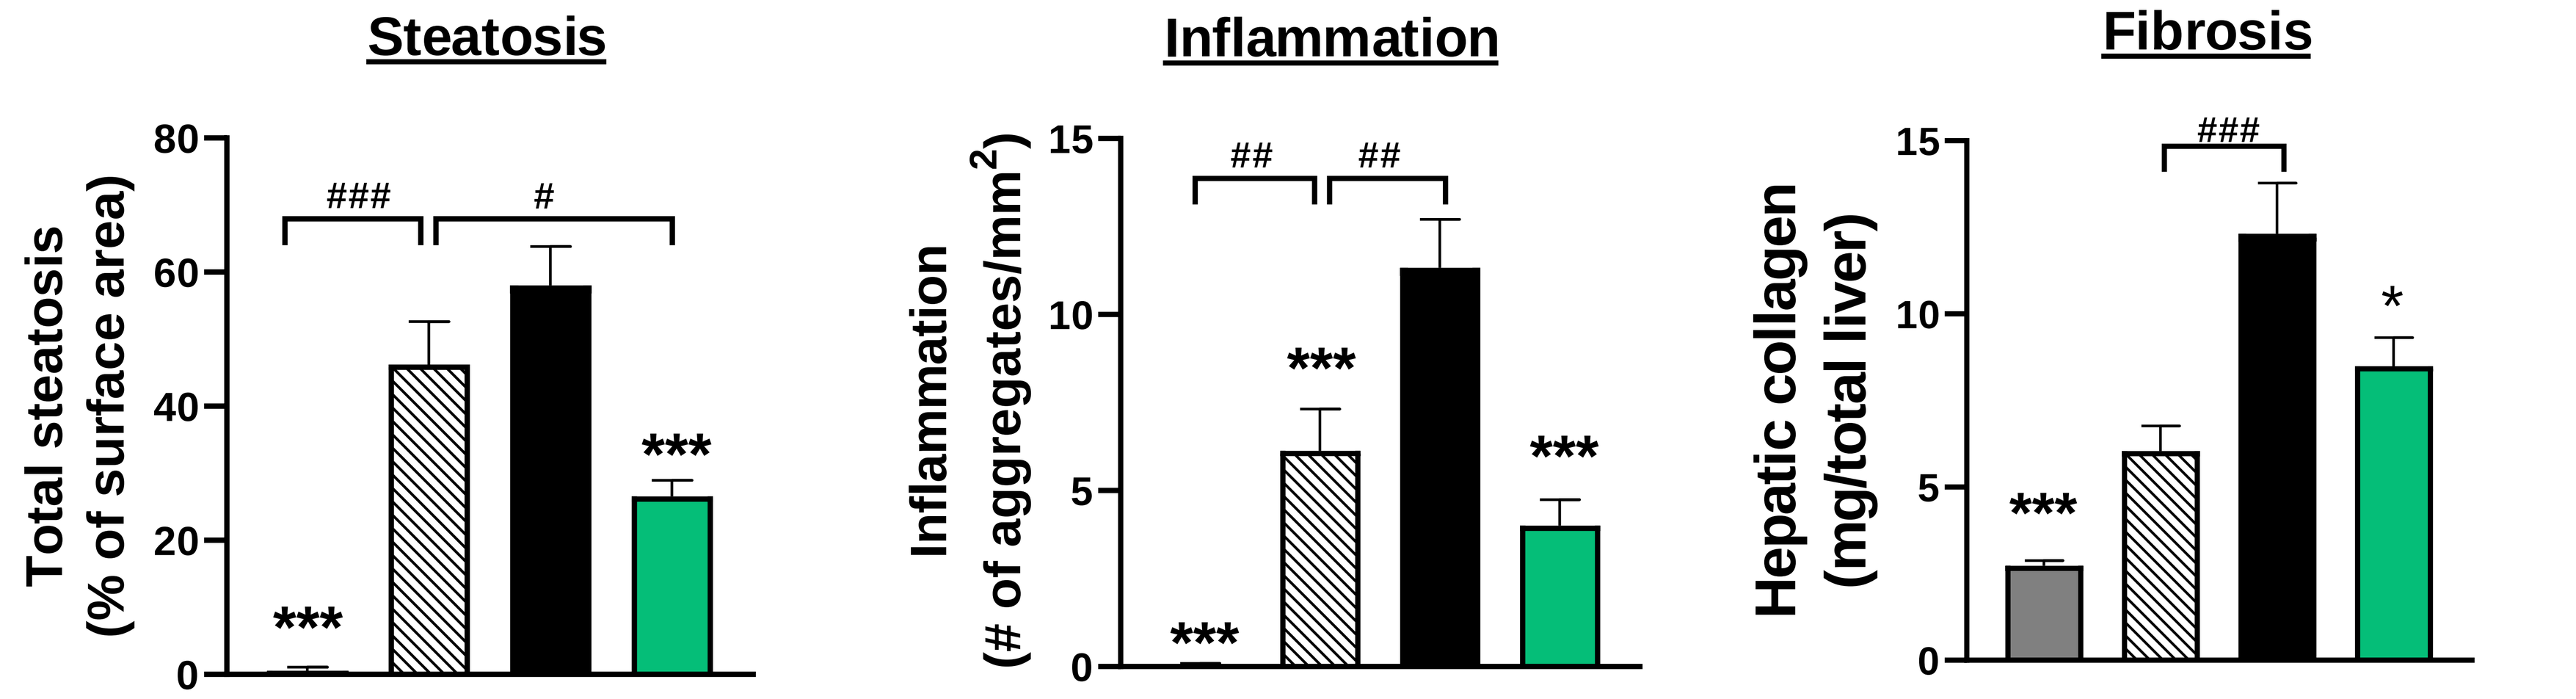
Steatosis
Total steatosis (% of surface area)
Inflammation
Inflammation (# of aggregates/mm²)
Fibrosis
Hepatic collagen (mg/total liver)

All figures represent data mean + SD with *p<0.05, **p<0.01 and ***p<0.001 vs FFD-vehicle. *Chow n=8; vehicle, TVB-3664, n=14~15*

Conclusions

- Denifanstat has shown promise on LDL-C lowering and liver histological improvements in MASH patients in Phase 2b FASCINATE-2 study[2]
- FASN inhibitor not only reduced circulating cholesterol but also decreased the development of atherosclerosis and improved liver histology in a mouse model of dyslipidaemia and MASH
- FASN inhibitor also decreased inflammation by reducing several circulating inflammatory markers, such as CCL4 and CXCL2, in Ldlr-/- MASH mice
- These results suggest that a FASN inhibitor like denifanstat could provide benefit in cardiovascular as well as liver health, supporting clinical evaluation of denifanstat for long term outcomes in MASH patients

References

(1) O'Farrell et al., 2022. Scientific Reports. doi:10.1038/s41598-022-19459-z

(2) Loomba et al., 2024. The Lancet Gastroenterology & Hepatology. doi:10.1016/S2468-1253(24)00246-2

(3) Loomba et al. 2021. Gastroenterology. doi:10.1053/j.gastro.2021.07.025

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com